Beyond TurboTMT: Phi-SDM Super-resolution Methods for Next-generation Highly-multiplexed Quantitative Ultrasensitive and Single-Cell Proteomics via TMTPro Complement Ion Deconvolution

Hendrik Wesseling[1]; Konstantin Aizikov[2]; Dmitry Grinfeld[2]; Arne Kreutzmann[2]; Daniel Marc Mourad[2]; Oliver Lange[2]; Alexander A. Makarov[2]; Erik Hett[1]; David H. Perlman[1]

[1]MERCK Cambridge Exploratory Science Center, Cambridge, MA

[2]ThermoFisher Scientific (Bremen) GmbH, Bremen, Germany

Introduction

Single-Cell Proteomics has emerged as an extremely promising new field capable of unlocking the mechanisms behind the vast cellular heterogeneity that defines biology, disease, and therapeutic response. However, enormous challenges exist, including how to maximize both quantitative signal and experimental throughput. We utilize the multiplexed method (e.g., SCoPE-MS[1], SCoPE2[2,3]) in which individual cells' proteomes are labeled, pooled, and combined with abundant carrier material using tandem mass tag (TMT) reagents, improving both peptide identification and cell throughput. Still, entire proteomes must be gleaned through short single-shot nanoLC-MS runs that are plagued by limited ion current. Under these circumstances, relatively long trap fill-times are necessary to obtain sufficient peptide signal, and, optimally, both identification and accurate quantification need to be achieved from single MS2 fragmentation spectra for sufficient throughput. Unfortunately, accurate quantitation of TMT reporter ions in MS^2 remains challenging, due to the co-isolation artifacts they suffer.

TMT complement (TMTc) ions, consisting of the peptide plus TMT balancer moieties, have the potential to provide quantitative read-outs in the MS^2 spectra that are essentially free of co-isolation artifacts, so long as the ions can be sufficiently resolved. Specifically, in order to deconvolute the nearly isobaric ^{15}N and ^{13}C TMTc species of the TMTPro™ products so as to exploit these reagents' full multiplexing power, ultra-high spectral resolution is required.

Recently, Phi-SDM signal processing, which is capable of resolving TMT reporter ions from transients 2-3-times shorter than required by conventional eFT processing[4,5], was introduced and commercialized as the TurboTMT™ feature in the Orbitrap Eclipse™ and Exploris™ MS instrument lines. Phi-SDM is similarly capable of rendering 2-3-times higher resolution spectra than eFT for any given transient length. Here, we have explored whether Phi-SDM directed not at the TMT reporter ions, as in TurboTMT, but rather at the high-mass TMTc ions might provide sufficiently high-resolution measurement to enable their full deconvolution.

[1]Budnik, B., Levy, E., Harmange, G. et al. Mass spectrometry of single mammalian cells quantifies proteome heterogeneity during cell differentiation. *Genome Biol* 19, 161 (2018)
[2]Specht H, Emmott E, Petelski A, Huffman RG, Perlman DH, Serra M, Kharchenko P, Koller A, and Slavov N. Single-cell proteomic and transcriptomic analysis of macrophage heterogeneity using SCoPE2. *Genome Biol* volume 22, 50 (2021)
[3]Petelski A, Emmott E, Leduc A, Huffman RG, Specht H, Perlman DH, Slavov N. Multiplexed single-cell proteomics using SCoPE2. *Nature Protocols* (in press) (2021)
[4]Grinfeld D, Aizikov K, Kreutzmann A, Damoc E, and Makarov A. Phase-Constrained Spectrum Deconvolution for Fourier Transform Mass Spectrometry. *Anal. Chem.* 89, 2, 1202–1211. (2017)
[5]Kelstrup CD, Aizikov K, Batth TS, Kreutzman A, Grinfeld D, Lange O, Mourad D, Makarov A, Olsen JV. Limits for Resolving Isobaric Tandem Mass Tag Reporter Ions Using Phase-Constrained Spectrum Deconvolution. *J Proteome Res* 17 (11) , 4008-4016. (2018)

Methods

Single-Cell Proteomics dilution standards were made using commercial purified peptide standards (MassPREP™ Peptide Mixture, Waters; Pierce™ HeLa Digest/PRTC Standard, Thermo Scientific™; Mass Spec-Compatible Human Protein Extract, Promega) labeled with TMTPro™ reagents (Thermo Scientific), according to the SCoPE-MS paradigm. Samples were injected on the single-cell level and analyzed on a Q Exactive™ HF-X mass spectrometer (Thermo Scientific) outfitted with an Evosep™ Plus nano-flow LC device (Evosep), or an Orbitrap Exploris™ 480 mass spectrometer outfitted with a Vanquish™ Neo UHPLC system (Thermo Scientific). Research-grade versions of Tune instrument control software were employed that enabled high-resolution phase-constrained spectral deconvolution (Phi-SDM) across specified ranges, including TMTc ion regions. Data was acquired at various instrument resolution settings (as indicated), using either eFT or Phi-SDM processing, with ion fill-times adjusted to the maximum afforded for complete parallelization with transient acquisition. Spectra were visualized using Xcalibur™ software (Thermo Scientific).

Cellular Heterogeneity is Fundamental to Human Biology

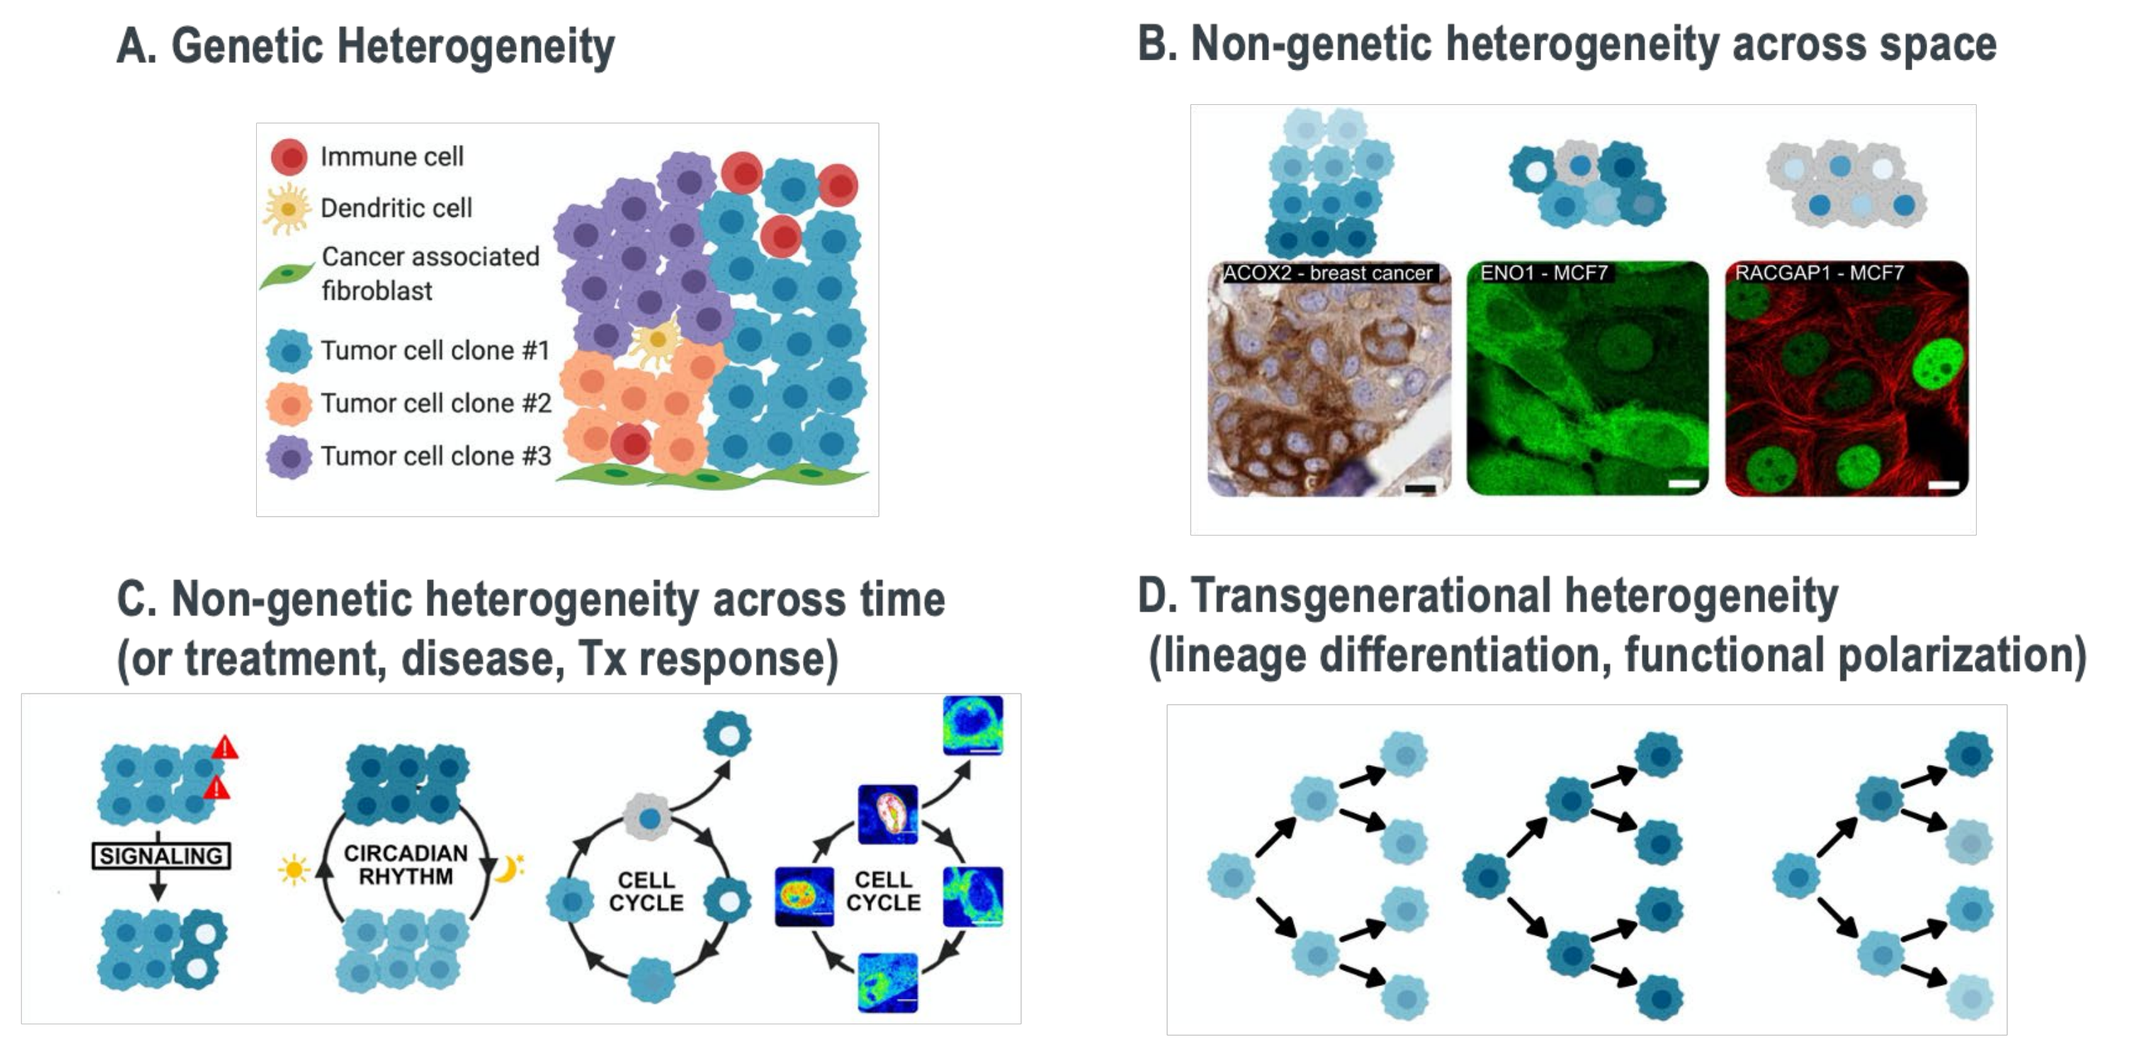

A. Genetic Heterogeneity
Immune cell
Dendritic cell
Cancer associated fibroblast
Tumor cell clone #1
Tumor cell clone #2
Tumor cell clone #3
B. Non-genetic heterogeneity across space
ACOX2 - breast cancer
ENO1 - MCF7
RACGAP1 - MCF7
C. Non-genetic heterogeneity across time (or treatment, disease, Tx response)
SIGNALING
CIRCADIAN RHYTHM
CELL CYCLE
CELL CYCLE
D. Transgenerational heterogeneity (lineage differentiation, functional polarization)

Mammalian cells are defined by their phenotypic and proteomic heterogeneity. This heterogeneity can be classified into at at lease 4 separate categories: (A) Genetic heterogeneity, (B) Non-genetic heterogeneity across space, (C) Non-genetic heterogeneity across time, and (D) Transgenerational heterogeneity.

Gnann C, Cesnik AJ, and Lundberg E., *Trends in Cancer*, Volume 7, Issue 4, P278-282, April 01, 2021.

The SCoPE-MS Method For Multiplexed Single-Cell Proteomics

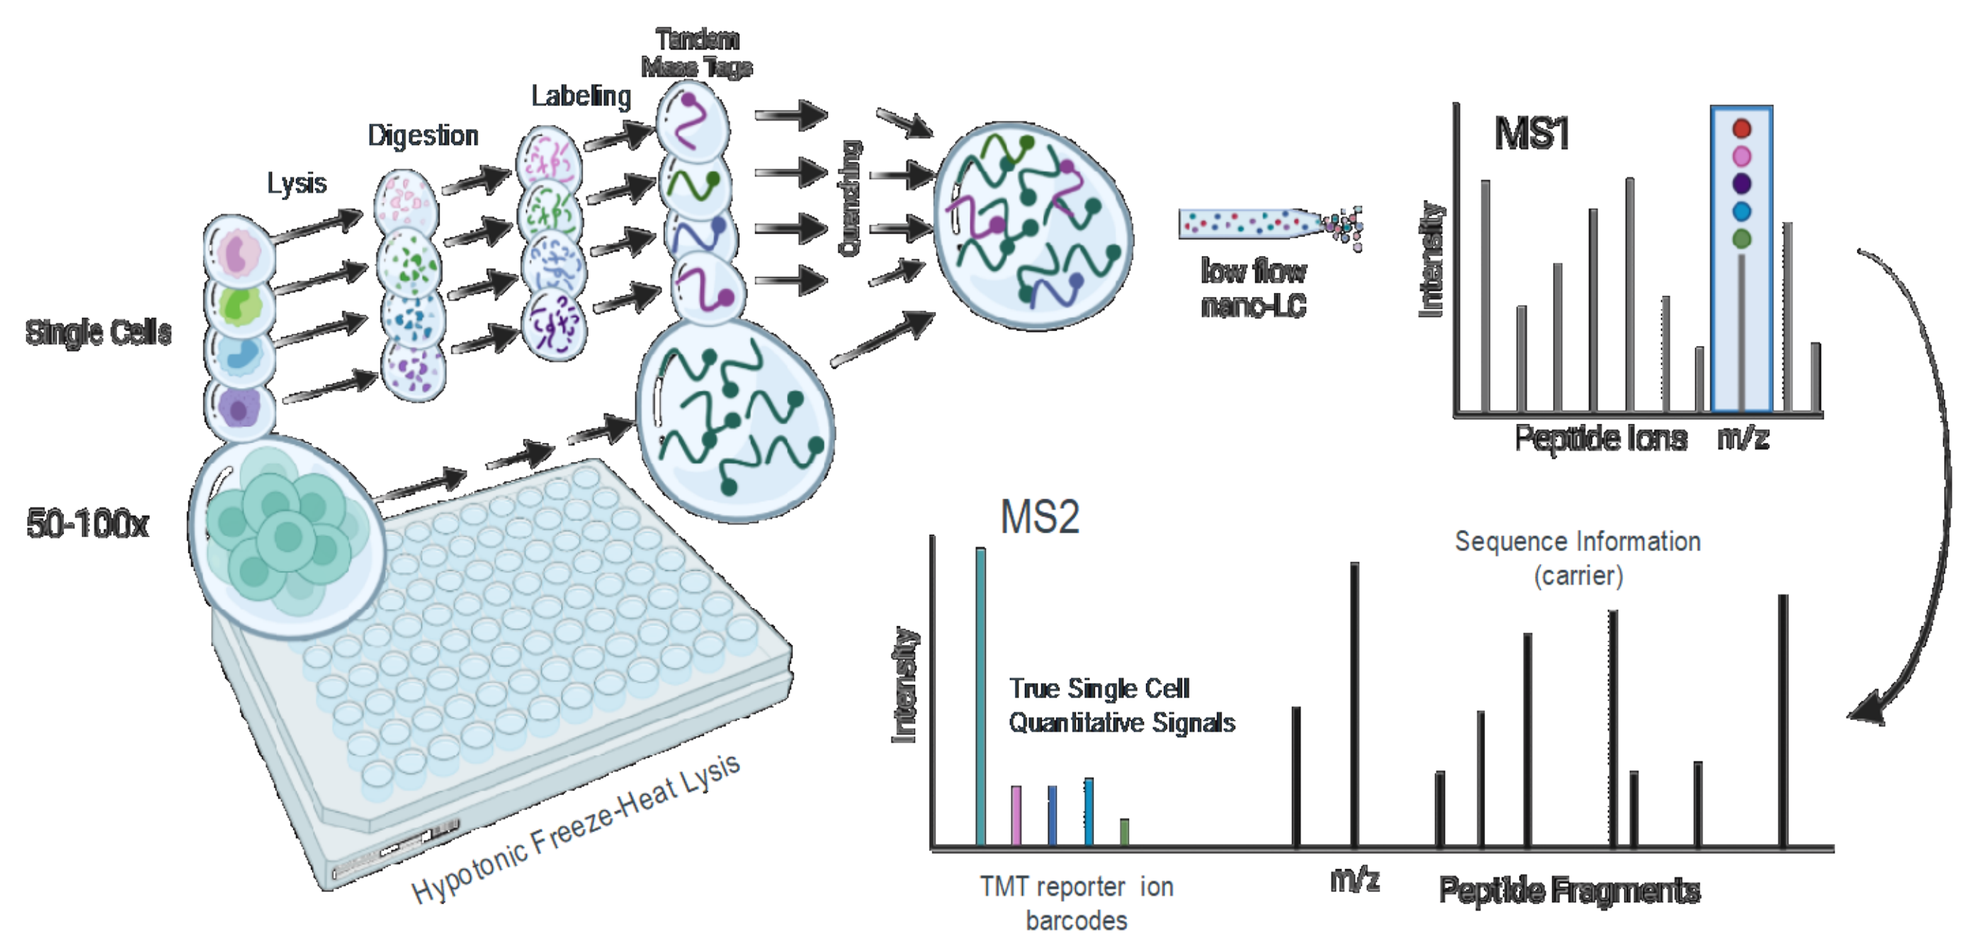

Single Cells
50-100x
Lysis
Digestion
Labeling
Tandem Mass Tags
Quenching
low flow nano-LC
MS1
Intensity
Peptide Ions
m/z
MS2
Sequence Information (carrier)
True Single Cell Quantitative Signals
TMT reporter ion barcodes
Peptide Fragments
Hypotonic Freeze-Heat Lysis

In the SCoPE-MS method for Single-Cell Proteomics1-3, single mammalian cells are subject to a mass-spec compatible lysis free of harsh detergents or chaotropes, followed by digestion, TMT labeling, and pooling with an abundant (e.g., 20-200 cell-equivalent) TMT-labeled carrier material. Upon LC-MS/MS of the pooled sample, fragmentation spectra contain abundant peptide backbone ion signals lent to the sample from the carrier, which facilitate identification, and also minute, authentic single-cell TMT reporter ion signals that read out relative peptide abundance in each single cell.

Challenging Features of Single-Cell Proteomic LC-MS

- **Single shot LC-MS**
 - "All your eggs in one basket": demands highest chromatographic resolution and extreme frugality to achieve maximal analytical depth.
- **Signal starved**
 - Majority of ions require long fill-times (e.g., 256ms, 512ms, etc.) for each MS2 scan.
 - *Coordinates well with longer transient lengths (e.g., 120K res, 240k res settings) for high resolution Orbitrap measurement.*
- **Duty-cycle constraints**
 - Due to time pressure and signal-intensity constraints, identification and quantification must be accomplished in MS^2. MS^3-based methods provide diminishing returns.
- **Need to maximize cell throughput**
 - Necessitates maximal multiplexing, short LC runs, etc.
- **TMT reporter ion species suffer MS^2 co-isolation artifacts**
 - Caused by the presence of isobaric peptides, TMT-quench products (1+ and multiply charged), and TMT-labeled protein material (semi-intact trypsin, etc.)
 - Long fill times increase opportunity for co-isolation

TMT Complement Ions for Accurate Multiplexed Quantitation

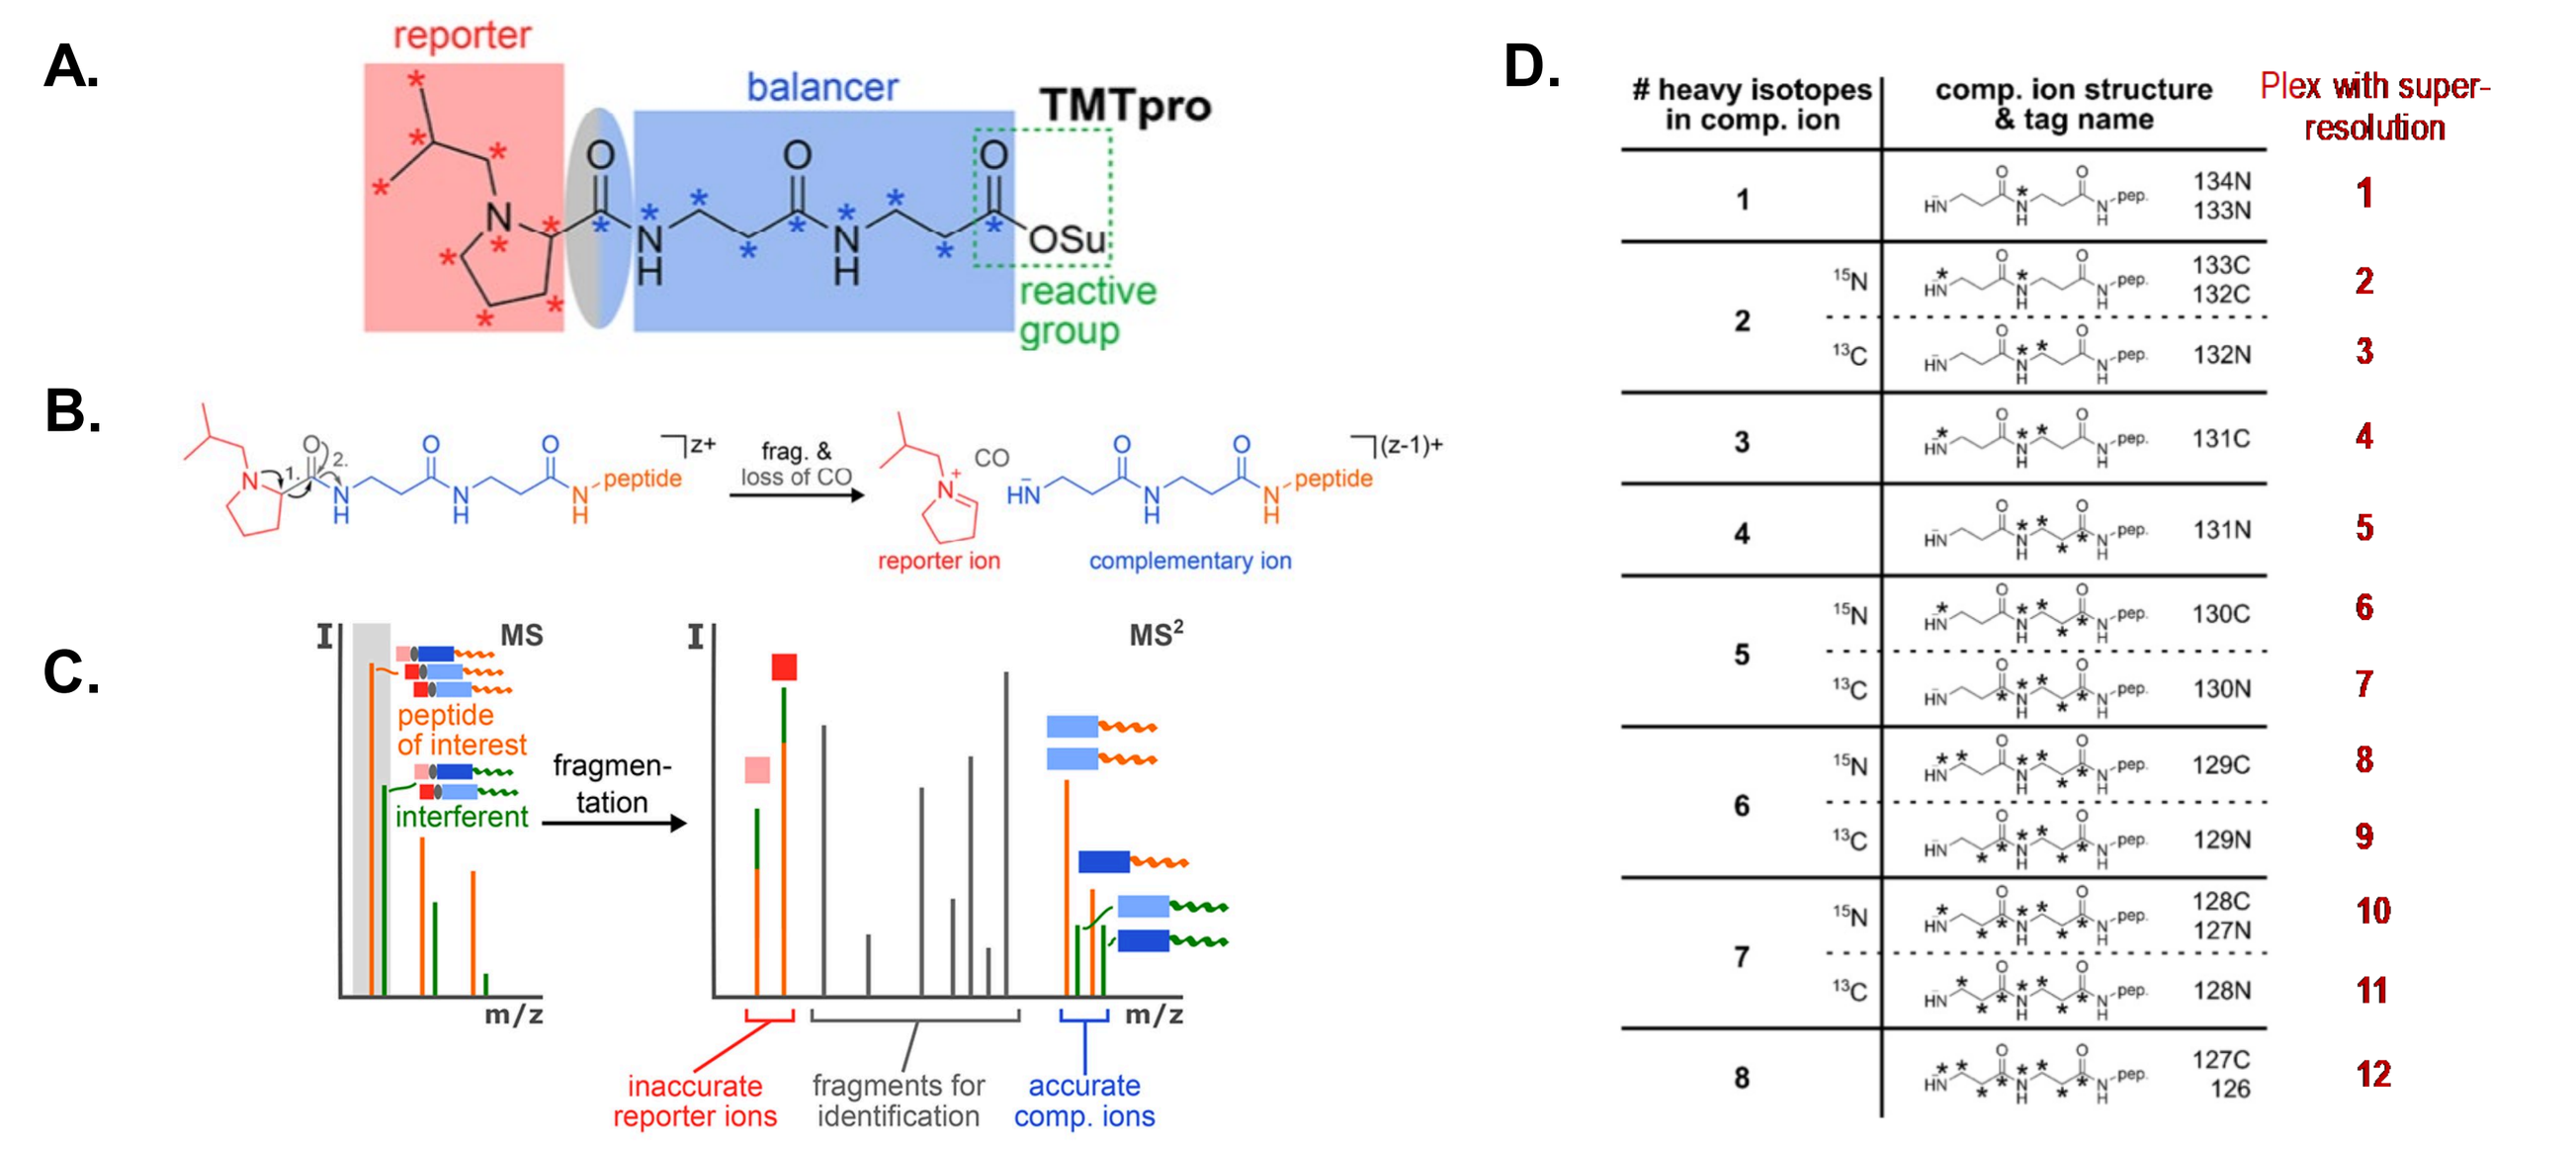

A.
reporter
balancer
TMTpro
OSu
reactive group
B.
peptide
frag. & loss of CO
reporter ion
complementary ion
C.
MS
peptide of interest
interferent
fragmen-tation
MS²
m/z
inaccurate reporter ions
fragments for identification
accurate comp. ions
D.
heavy isotopes in comp. ion
comp. ion structure & tag name
Plex with super-resolution
1 134N 133N 1
2 15N 133C 132C 2
13C 132N 3
3 131C 4
4 131N 5
5 15N 130C 6
13C 130N 7
6 15N 129C 8
13C 129N 9
7 15N 128C 127N 10
13C 128N 11
8 127C 126 12

Quantitation via TMTc. (A) The structure of the TMTPro family of labels consisting of the reporter moieties joined to the balancer regions. (B) During gas-phase fragmentation and TMT-RI formation, the TMTc ion-species is simultaneously formed from the TMTPro balancer moiety fused to the parent peptide, thus it has a peptide-specific, plus TMT-tag specific mass. (C) When a TMT-labeled peptide of interest (red, MS) is co-isolated along with an interferent (green), it produces a fragmentation spectrum (MS^2) in which the TMT-RIs from both the peptide and interferent are coincident, while their TMTc ion species retain distinct masses and are distinguishable. (D) The TMTPro 16-plex family forms 8 nominal mass TMTProC species, which can be further separated into 12 distinct species with ultra high-resolution methods.

Johnson, Stadlmeier & Wuhr, 2021

Detection of TMTPro Complement Ions in labeled Peptide STD

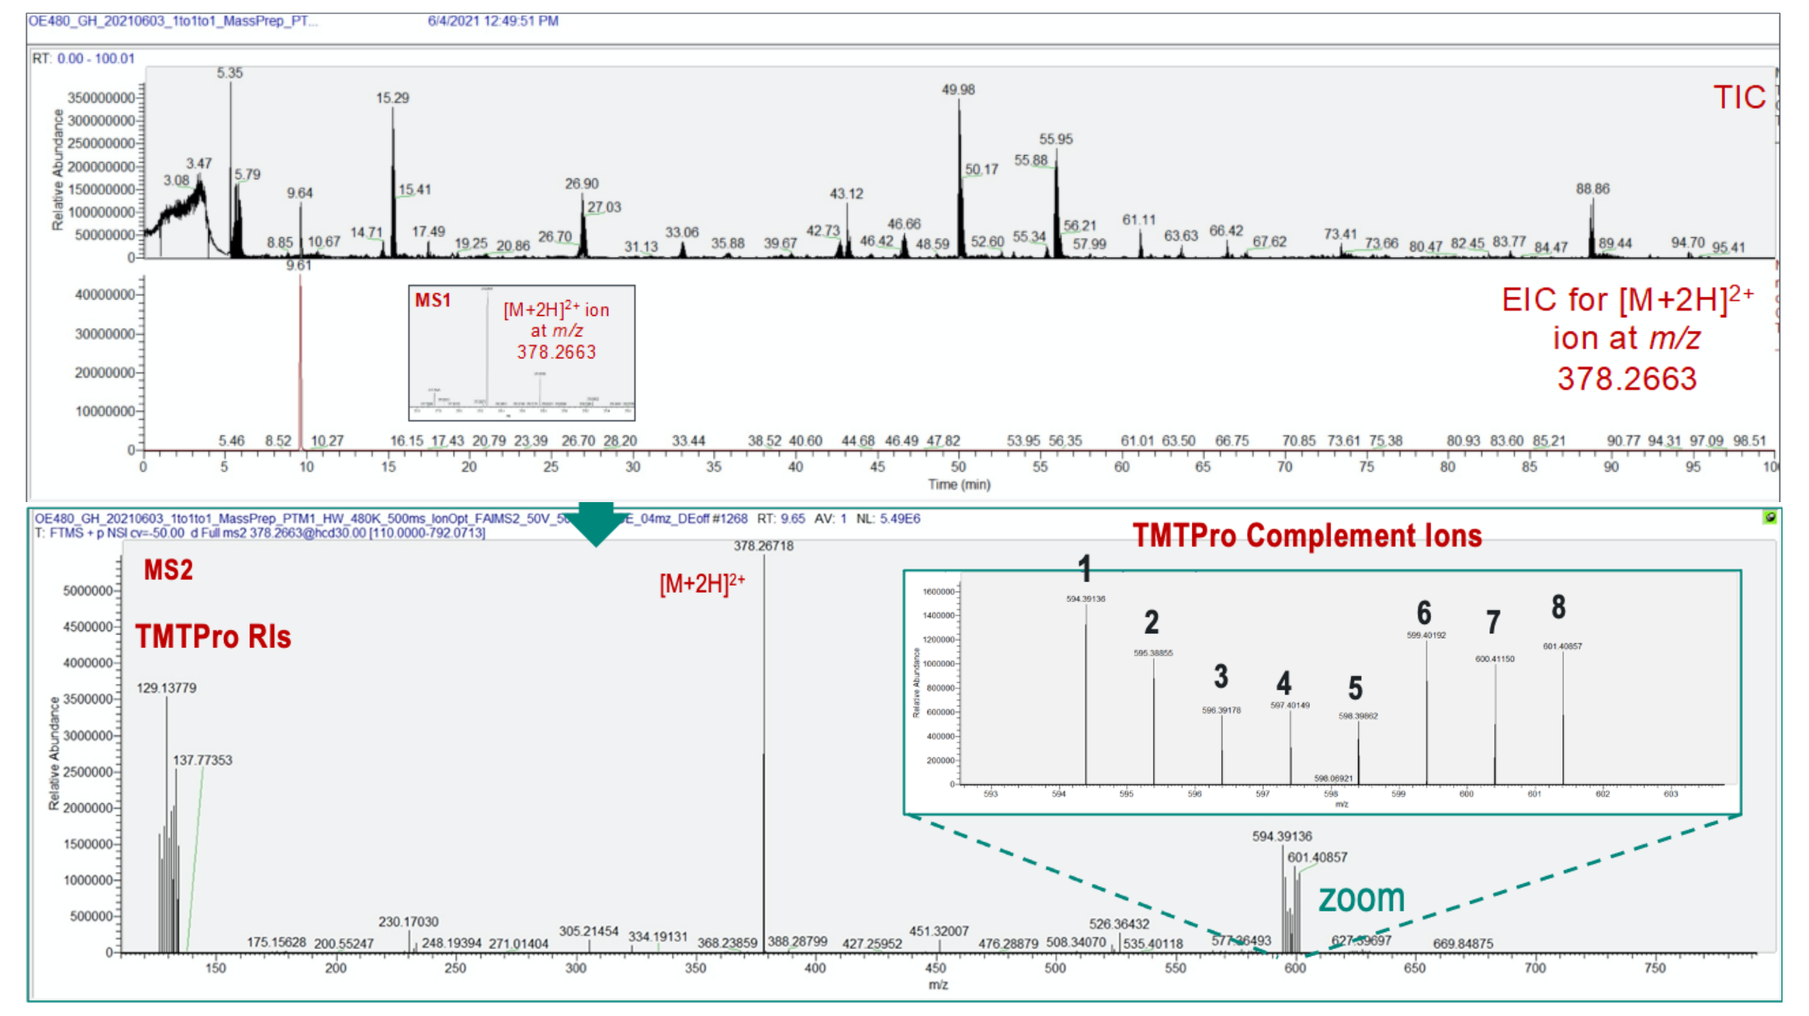

TIC
EIC for [M+2H]2+ ion at m/z 378.2663
MS1
MS2
TMTPro RIs
[M+2H]2+
TMTPro Complement Ions
1
2
3
4
5
6
7
8
zoom

LC-MS/MS of TMTPro labeled peptide standards yields MS^2 spectra displaying both low-mass TMTPro-RI species, and also prominent TMTProC ion clusters in the higher mass region. Zooming in on these TMTProC peaks show that they consist of the predicted 8 nominal mass species.

Insufficient Resolution of TMTProC Ions at 240k eFT Setting

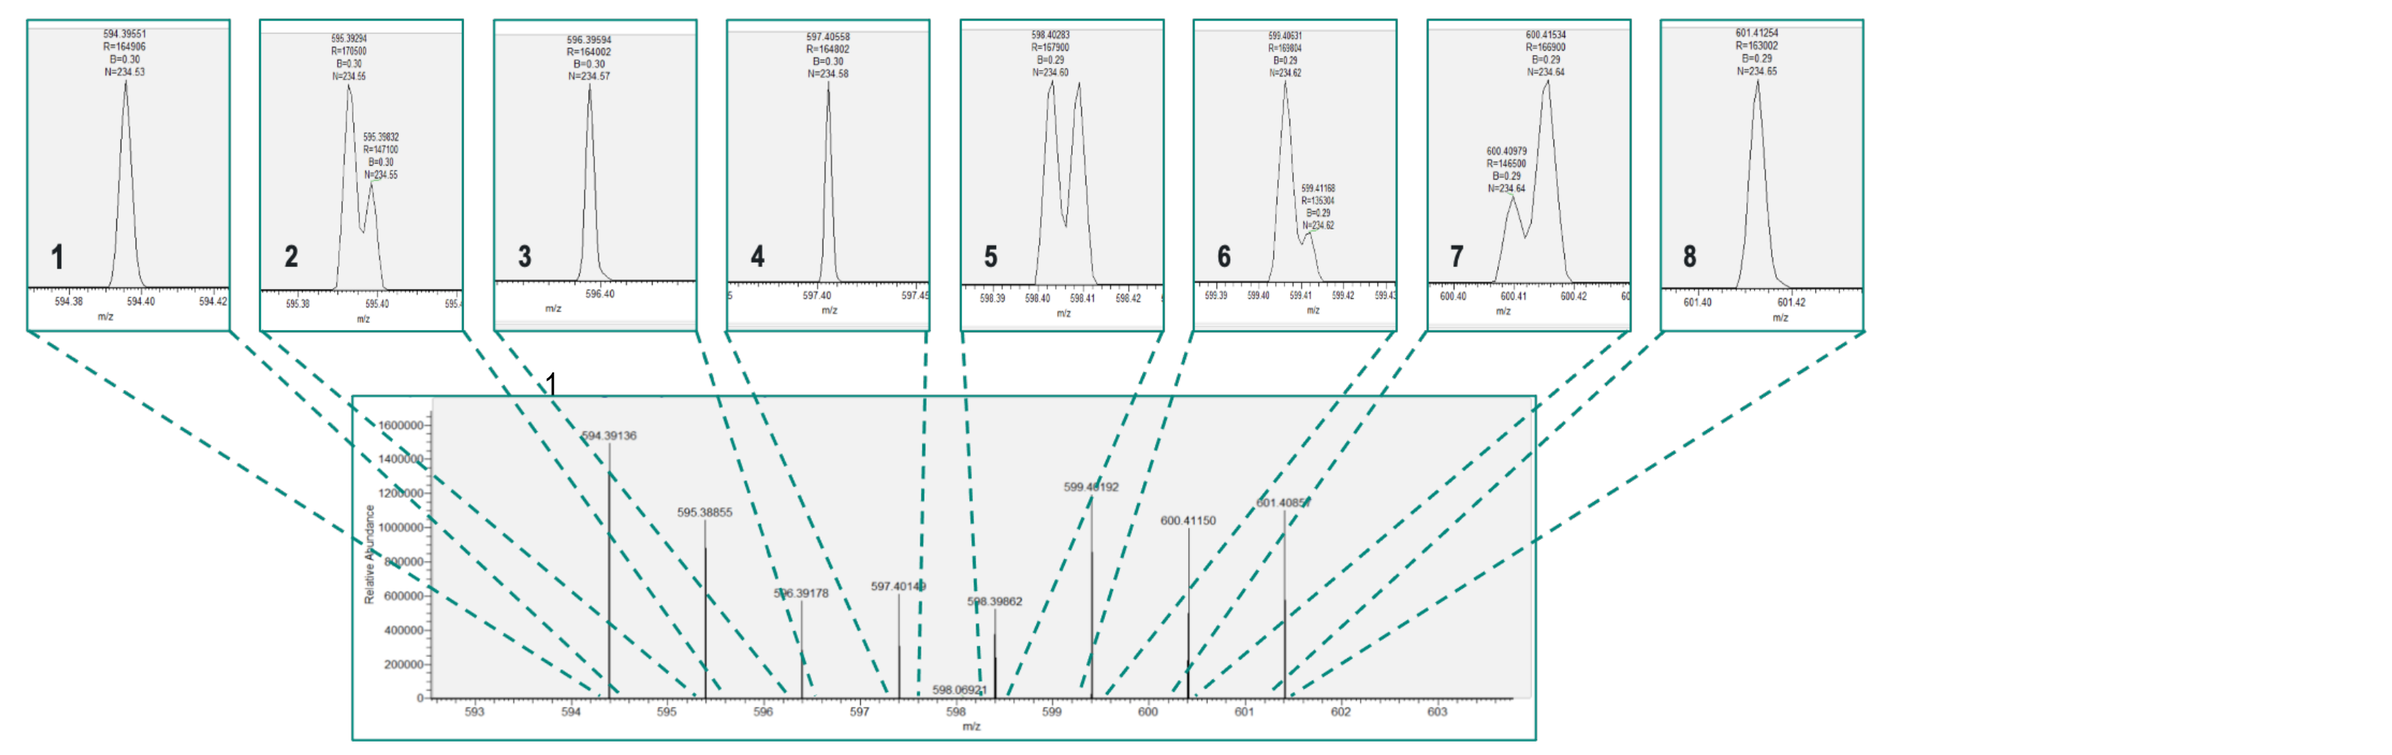

At the maximum resolution setting of the commercial Q Exactive HF-X instrument (240K using eFT) we are unable to completely resolve the 12 TMTProC species from the 8 nominal mass peaks. Zoomed in regions around each ion are shown above the 8-peak TMTProC cluster spectrum.

PhiSDM TMTProC Ion Deconvolution from a 512ms Transient

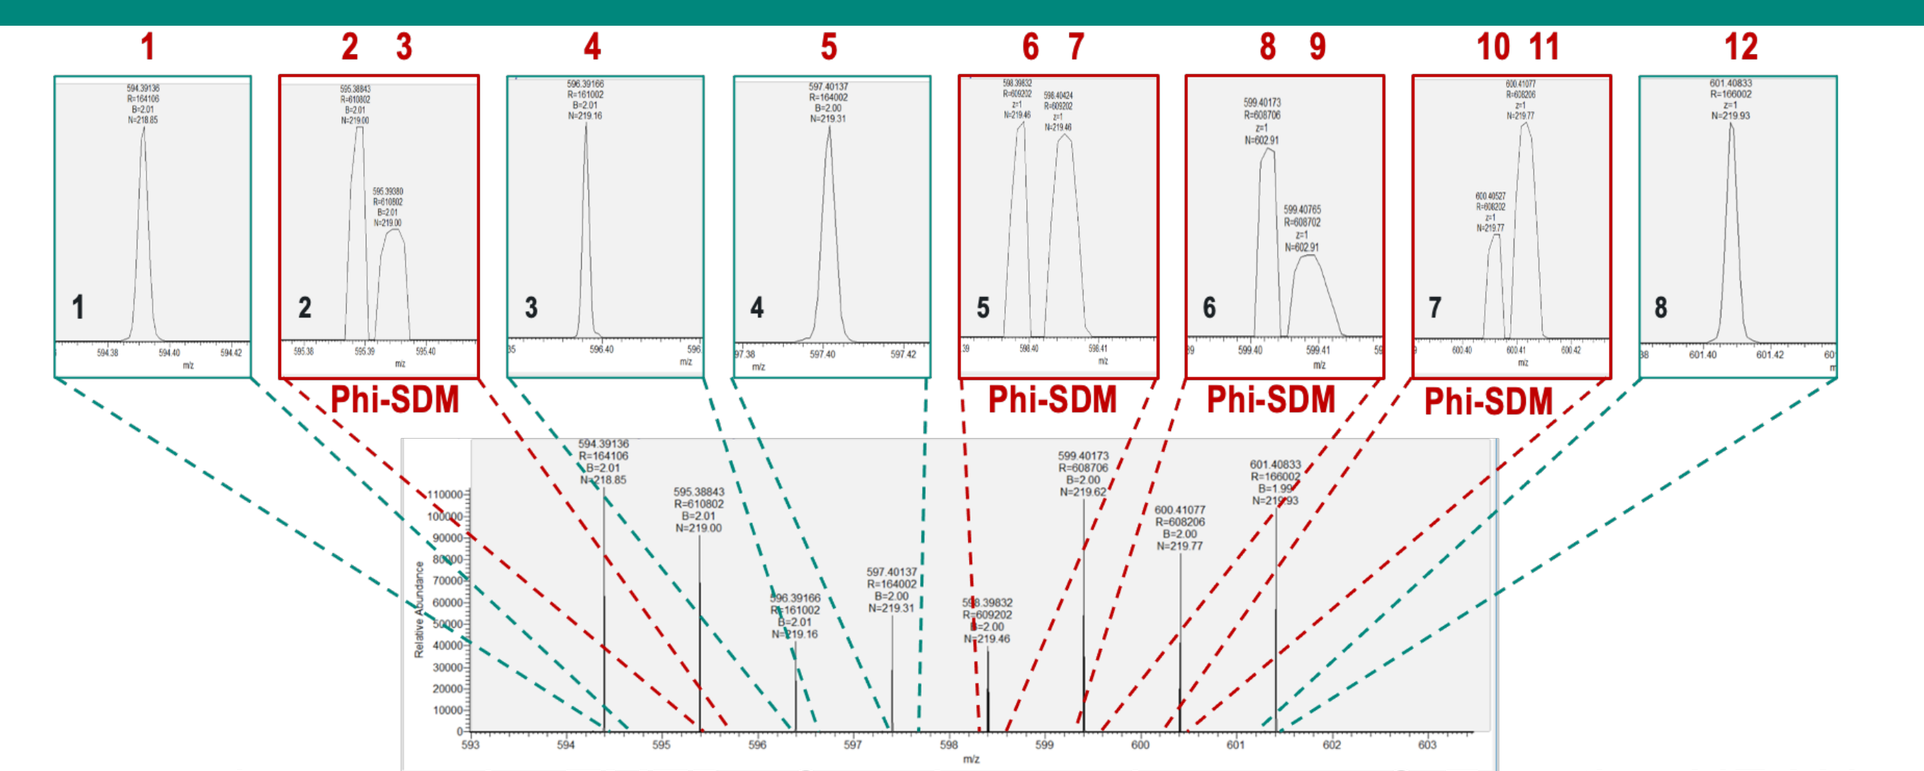

1
2 3
4
5
6 7
8 9
10 11
12
Phi-SDM
Phi-SDM
Phi-SDM
Phi-SDM

Enabling phi-SDM processing over the TMTProC region on the same Q Exactive HF-X instrument using the same transient acquisition settings as above (240K), allows us to completely resolve all 12 TMTProC species. Estimates of the empirical resolution at the peaks subjected to phi-SDM are >600K, while the resolution at the neighboring peaks subjected to eFT is ~160K

Sufficient Peak Calling with PhiSDM of a 256ms Transient

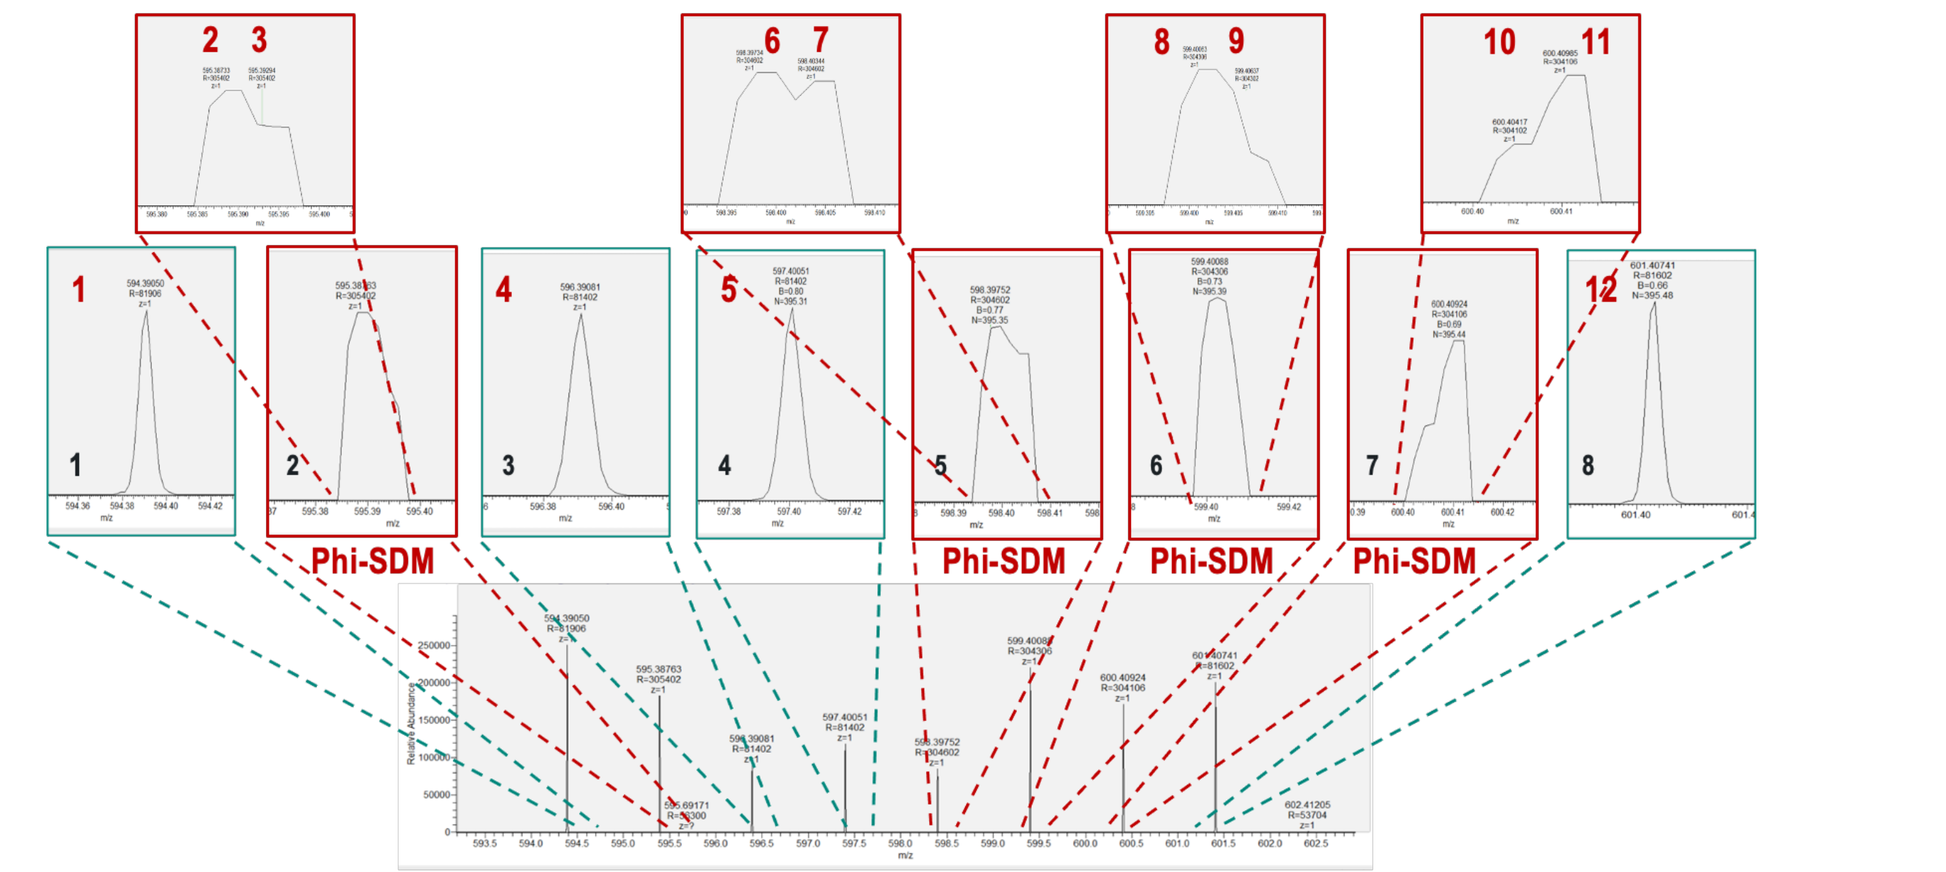

2 3
6 7
8 9
10 11
1
4
5
12
Phi-SDM
Phi-SDM
Phi-SDM
Phi-SDM

Reducing the transient length by half (nominally a 120K setting) and performing phi-SDM over the TMTProC regions as above, yields incompletely resolved peaks with partial splitting, at least as rendered by Xcalibur. However, zooming into these peaks shows that the phi-SDM algorithm was indeed able to identify and call out both of the peaks in each case.

Design of SCoPE-style Single Cell Standards

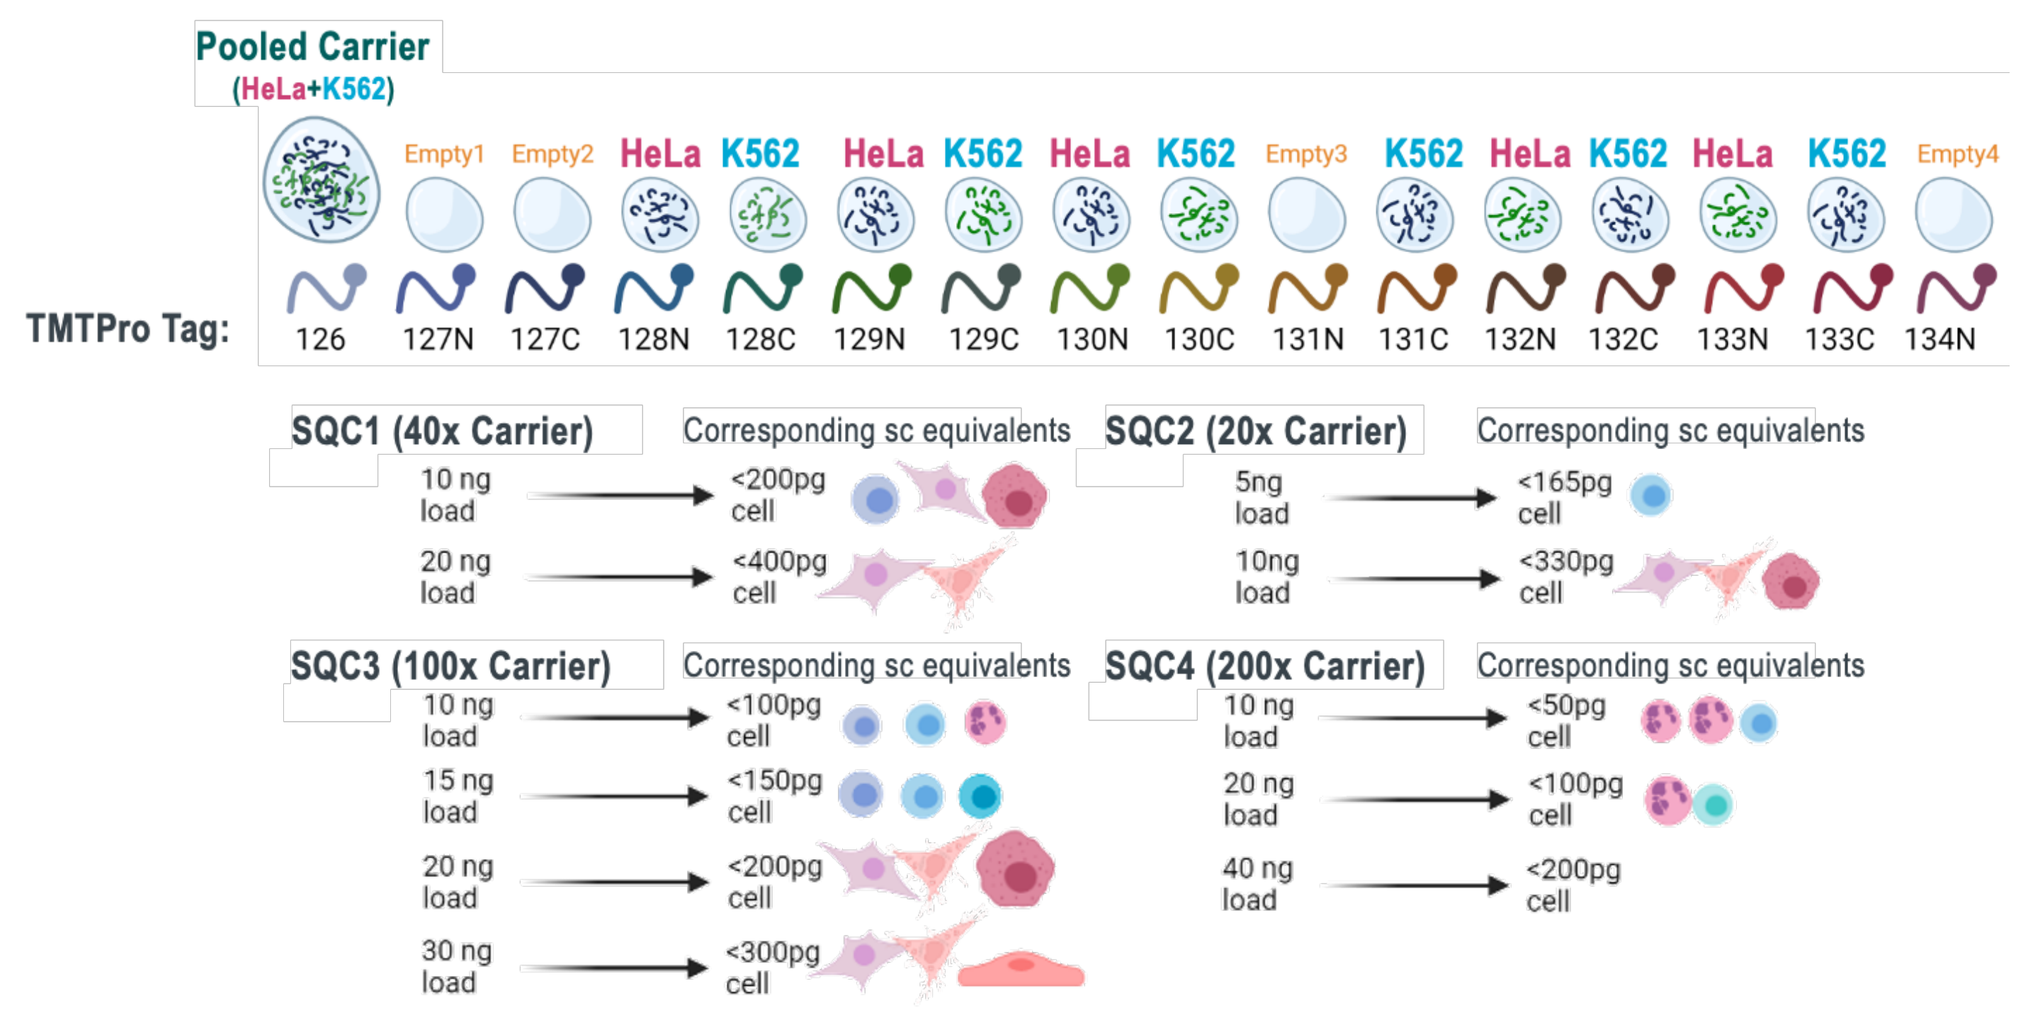

Pooled Carrier (HeLa+K562)
Empty1 Empty2 HeLa K562 HeLa K562 HeLa K562 Empty3 K562 HeLa K562 HeLa K562 Empty4
TMTPro Tag:
126 127N 127C 128N 128C 129N 129C 130N 130C 131N 131C 132N 132C 133N 133C 134N
SQC1 (40x Carrier)
10 ng load → <200pg cell
20 ng load → <400pg cell
SQC2 (20x Carrier)
5ng load → <165pg cell
10ng load → <330pg cell
SQC3 (100x Carrier)
10 ng load → <100pg cell
15 ng load → <150pg cell
20 ng load → <200pg cell
30 ng load → <300pg cell
SQC4 (200x Carrier)
10 ng load → <50pg cell
20 ng load → <100pg cell
40 ng load → <200pg cell
Corresponding sc equivalents

A set of SCoPE-style Single-Cell Proteomics QC standards (SQC) was designed to contain alternating peptides from 2 different cell types (HeLa and K562) in the single-cell TMT channels, along with a pooled carrier from the 2 cell types, and empty channels (to assess carryover). Different variants of the standards (SQC1-SQC4) were formulated to contain different ratios of carrier peptides to single cell channel peptides, from 20x to 200x, such that with a 10ng SQC sample load we could model the proteomic yield of different sizes of cells in the single cell channels.

Resolution and Quantification of Single Cell STD TMTProC Ions

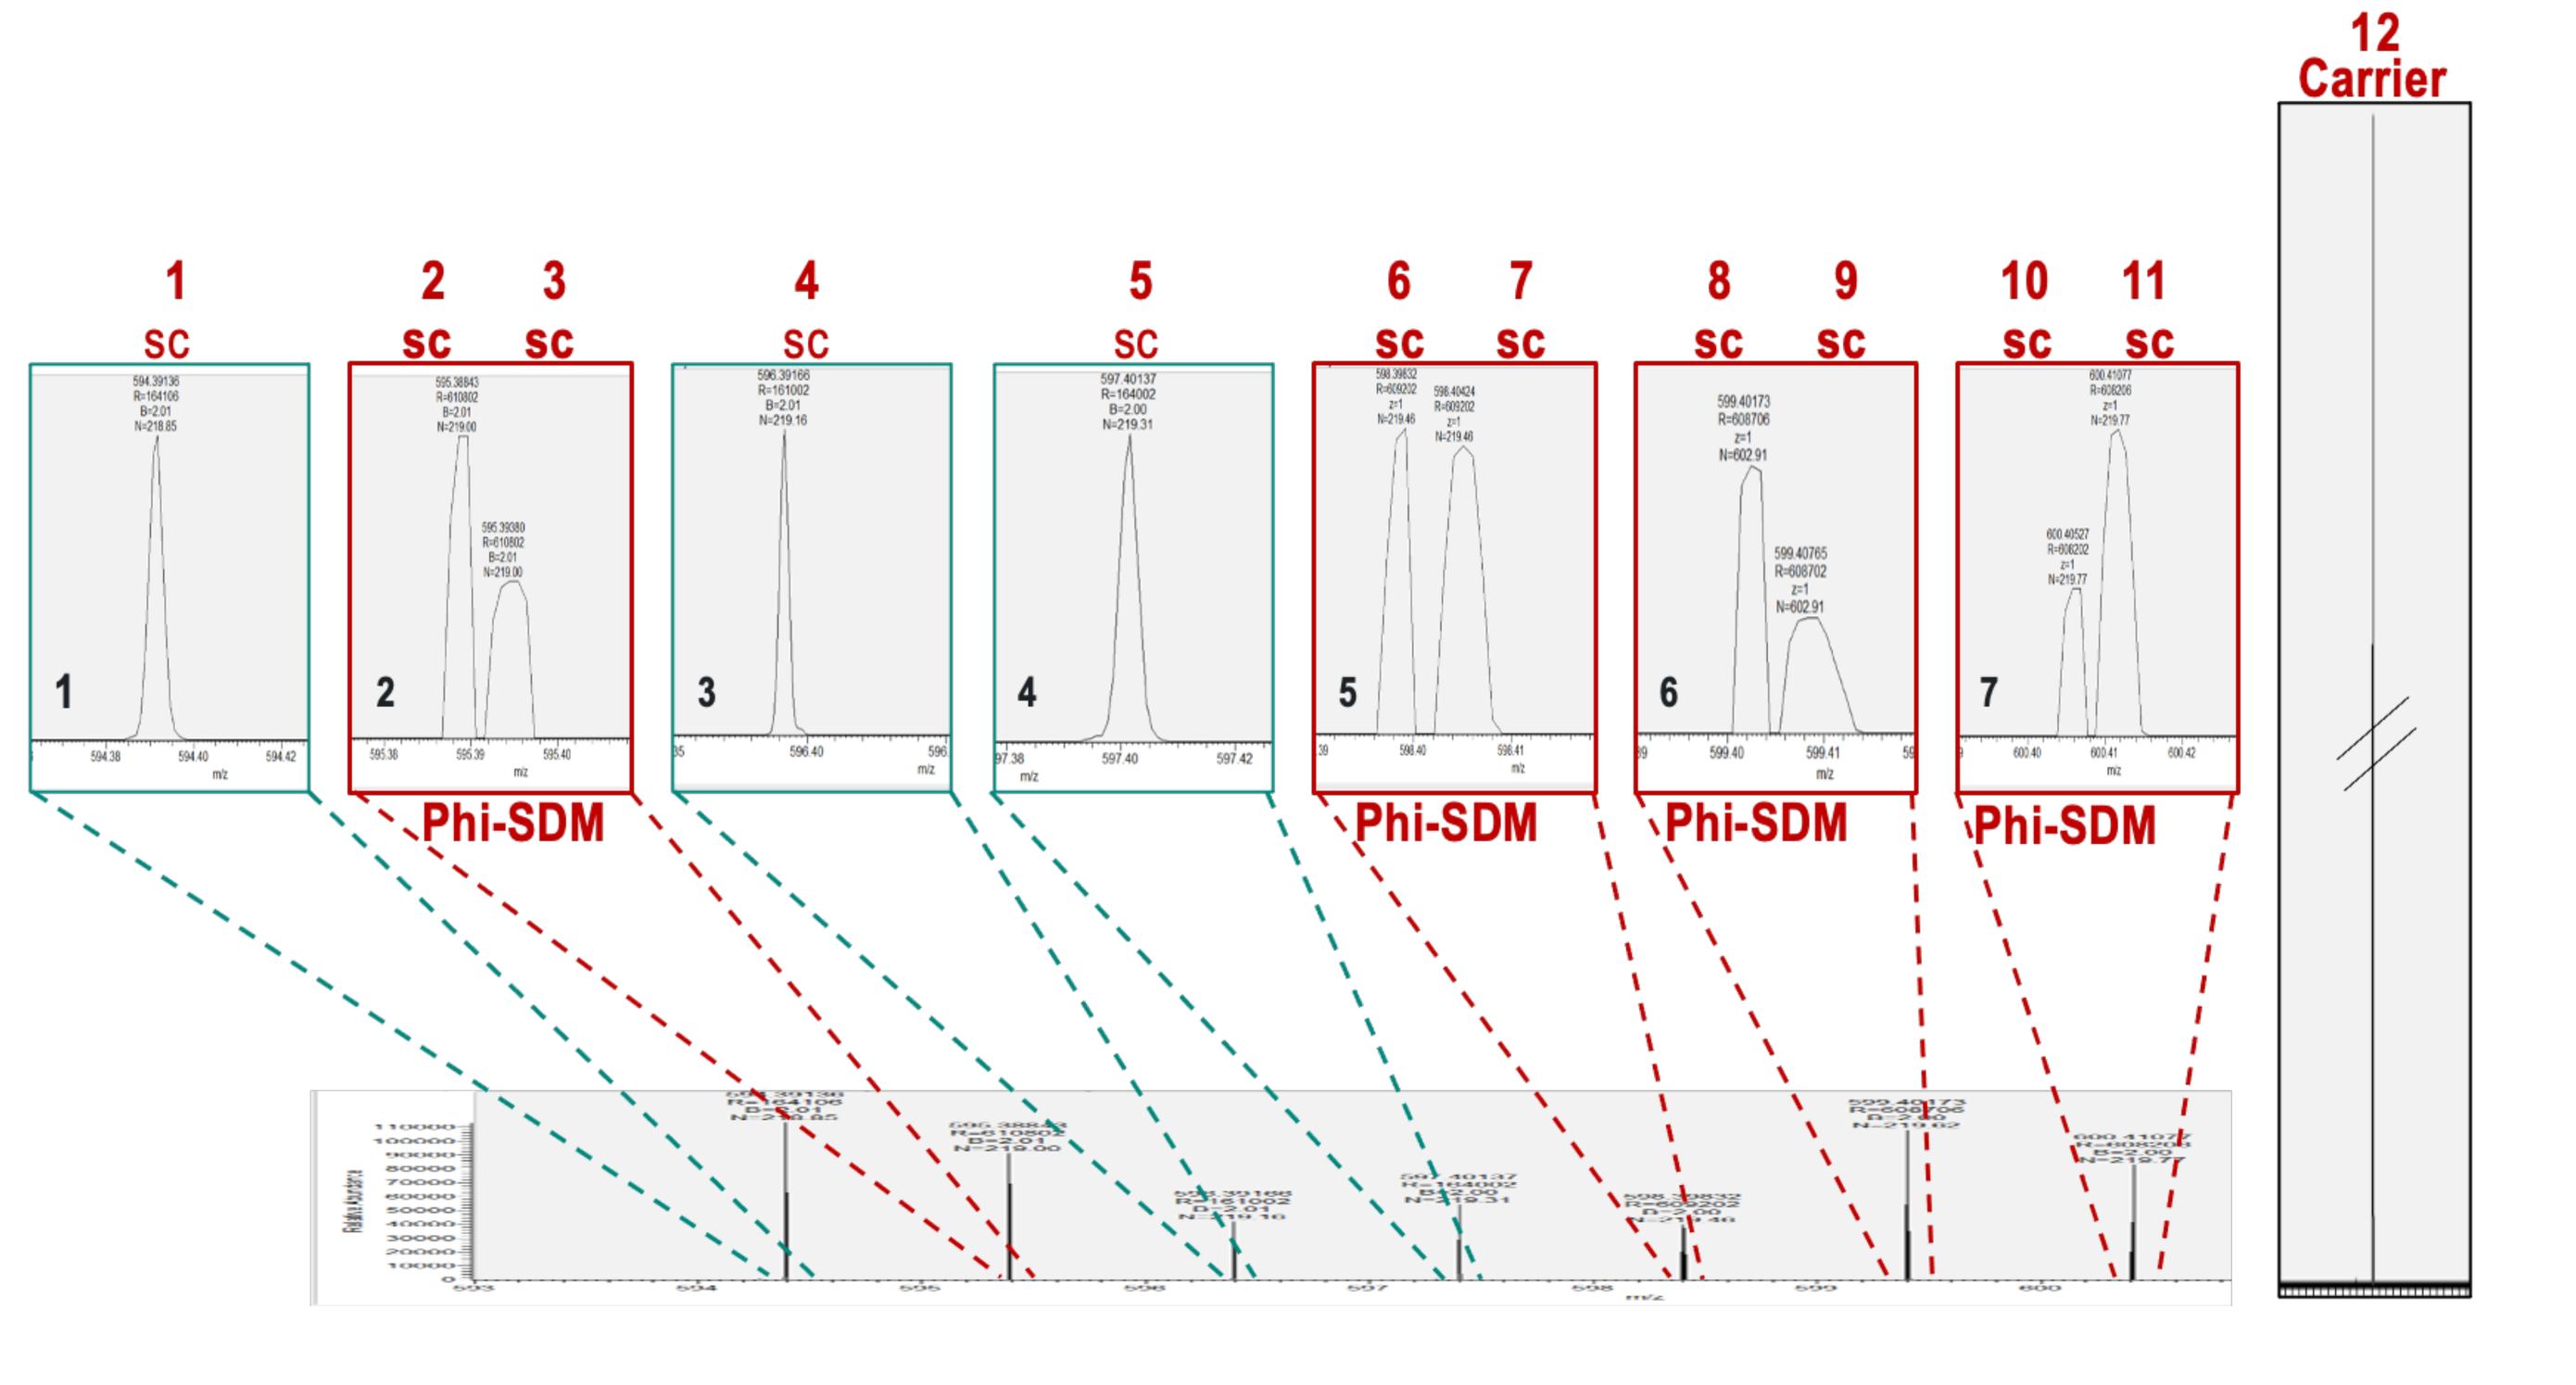

12 Carrier
1 SC
2 SC 3 SC
4 SC
5 SC
6 SC 7 SC
8 SC 9 SC
10 SC 11 SC
Phi-SDM
Phi-SDM
Phi-SDM
Phi-SDM

We are able to resolve and quantify each of the 12 TMTProC ions from our Single-Cell Proteomic SQC samples using phi-SDM processing across the TMTProC region on an Exploris 480 instrument. Notably the 126-labeled carrier (Peak 12) is well separated from the lower-mass single-cell channel signals and does not interfere with them. Therefore, with this design, 10 single cells plus 1 empty control well could be incorporated into each multiplexed Single-Cell Proteomics set.

Summary

Here we demonstrate that Phi-SDM applied across mass ranges spanning TMTc ions present in the tandem mass spectra of TMTPro-labeled peptides and single-cell standards deliver effective resolution values across these regions that surpass any achievable on current commercially available Orbitrap platforms. Moreover, we demonstrate baseline resolution of the nearly isobaric 15N and 13C TMTc species (delta mass ca. 6.3 mDa) at transient lengths of ca. 512ms and 256ms, corresponding to the nominal 240K and 120K eFT resolution settings, which synchronize well with maximum ion-fill times commonly used during single-cell analyses. This novel application of Phi-SDM beyond TurboTMT is a demonstration of the potential of this super-resolution method and brings us closer to an operational workflow for TMTPro complement ion-based quantification in highly-multiplexed ultrasensitive and single-cell proteomics.

Copyright © 2021 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.